Briquilimab, an Anti-Human CD117 Antibody, Effectively Treats Cockroach Allergen-Induced Asthma Model in Mice Expressing Chimeric Human/Mouse CD117

Mang Yu, Sambidhan Kattel, Andrew Wells, Revati Nerkar, Karl Meneses, Hye-Sook Kwon, Wendy W. Pang
Jasper Therapeutics, Inc., Redwood City, CA, USA

Briquilimab can potently inhibit the signaling pathway of human stem cell factor (SCF) and CD117 (c-Kit), thereby regulating mast cell (MC) survival and functions in the context of allergic disorders, including allergic asthma and atopic dermatitis.

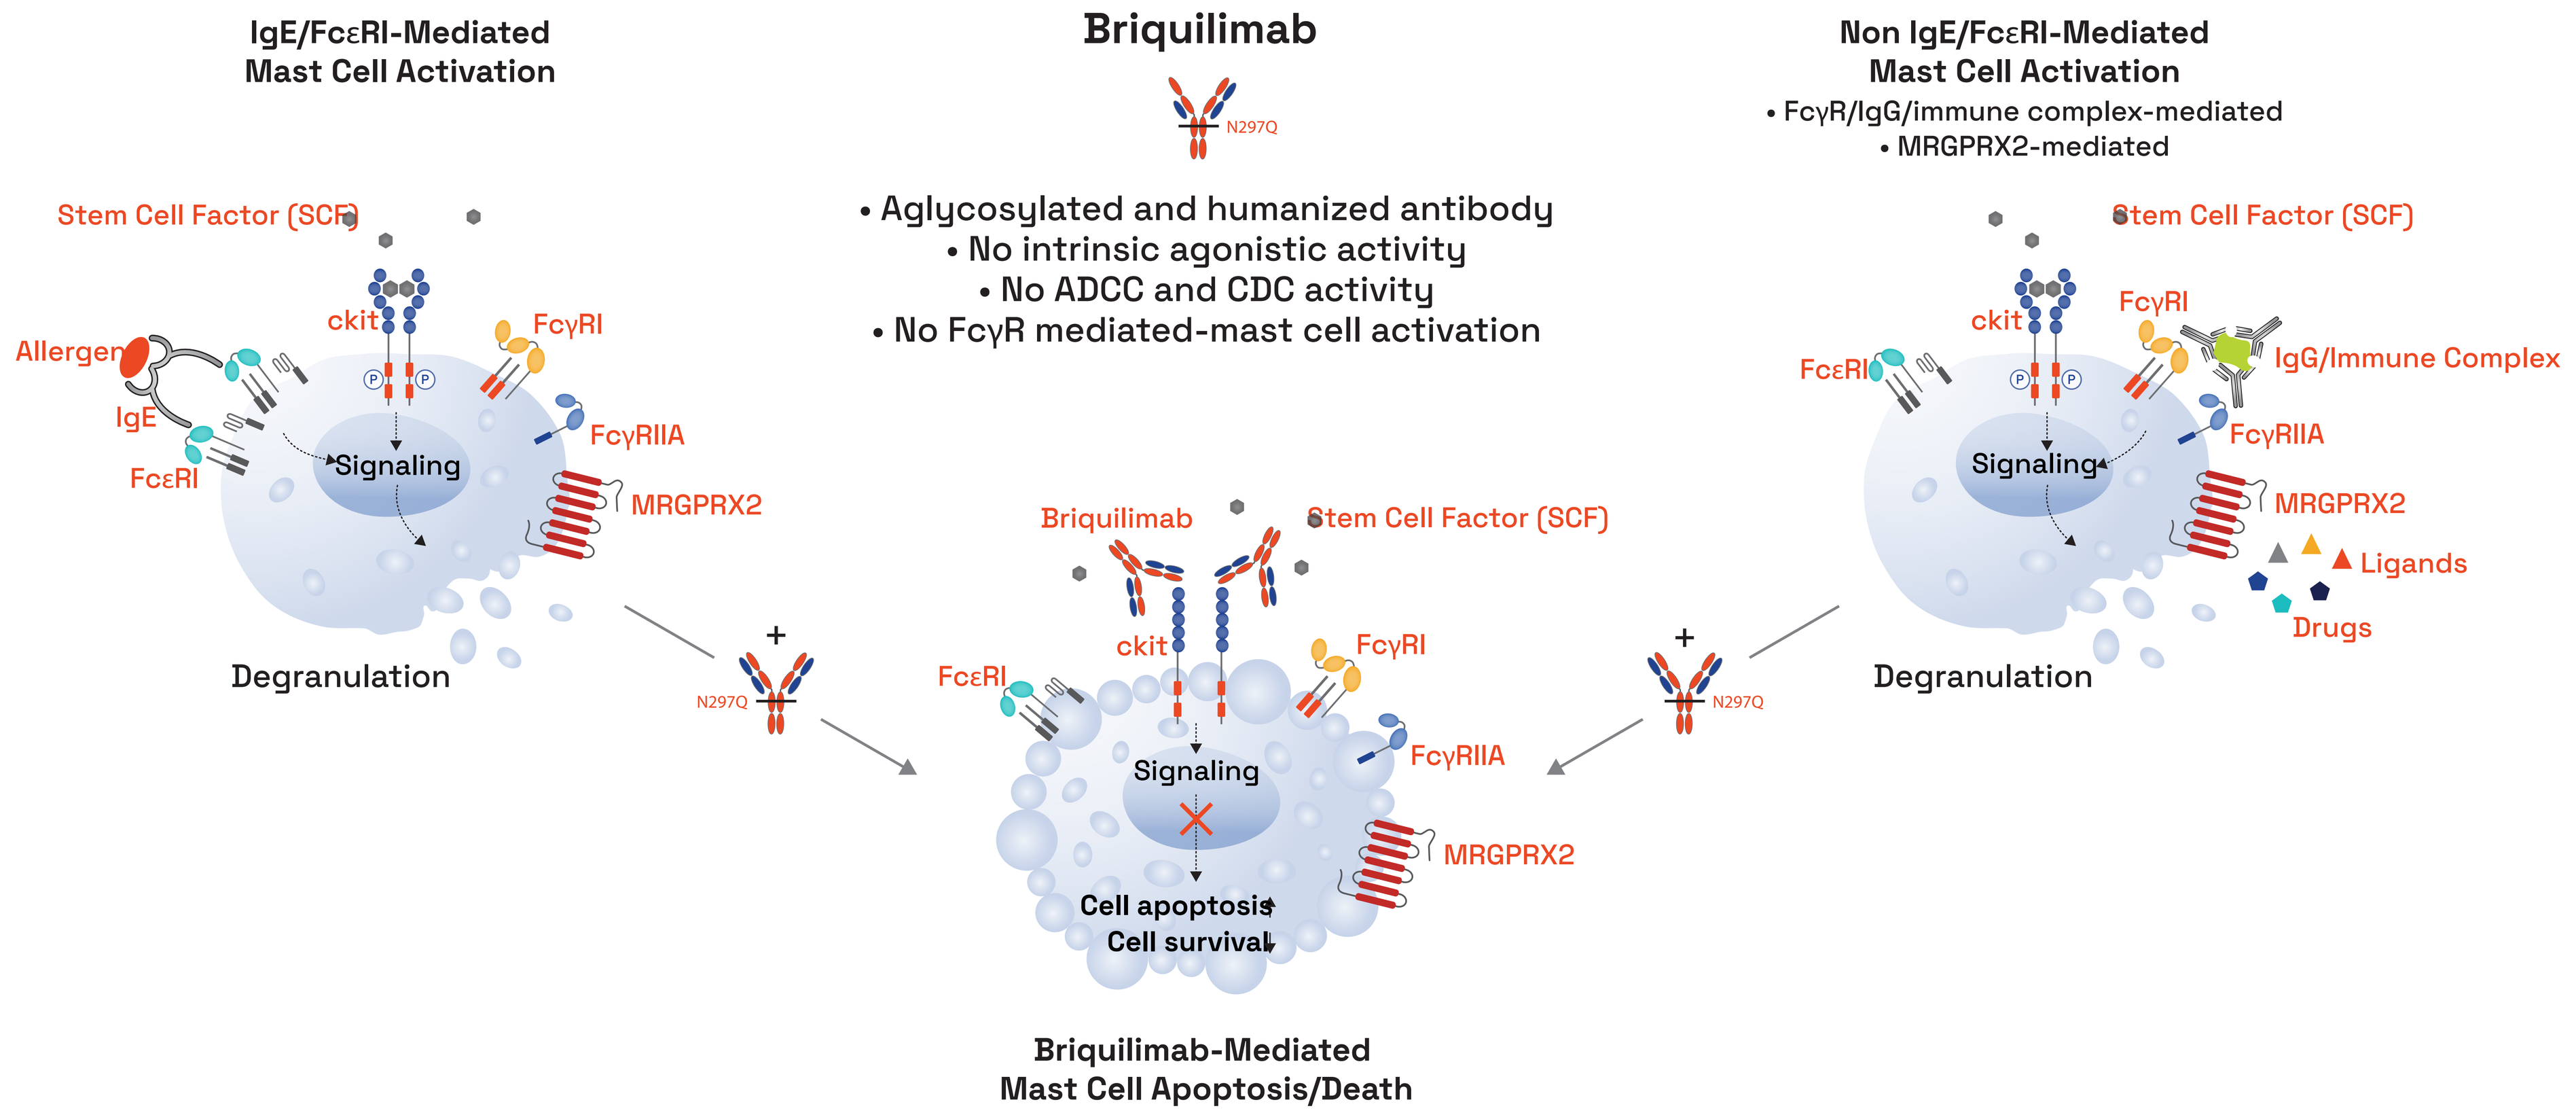
IgE/FcεRI-Mediated
Mast Cell Activation
Briquilimab
N297Q
• Aglycosylated and humanized antibody
• No intrinsic agonistic activity
• No ADCC and CDC activity
• No FcγR mediated-mast cell activation
Non IgE/FcεRI-Mediated
Mast Cell Activation
• FcγR/IgG/immune complex-mediated
• MRGPRX2-mediated
Stem Cell Factor (SCF)
ckit
FcγRI
Allergen
IgE
FcεRI
Signaling
FcγRIIA
MRGPRX2
Degranulation
IgG/Immune Complex
Ligands
Drugs
Briquilimab
Cell apoptosis
Cell survival
Briquilimab-Mediated
Mast Cell Apoptosis/Death

The transgenic h/mCD117 mice expressing both the functional human extracellular and murine intracellular domains of CD117 (c-Kit) enable proof-of-concept efficacy testing of briquilimab for allergic asthma by eliciting clinically relevant models in h/mCD117 mice following exposure to cockroach allergen (CRA).

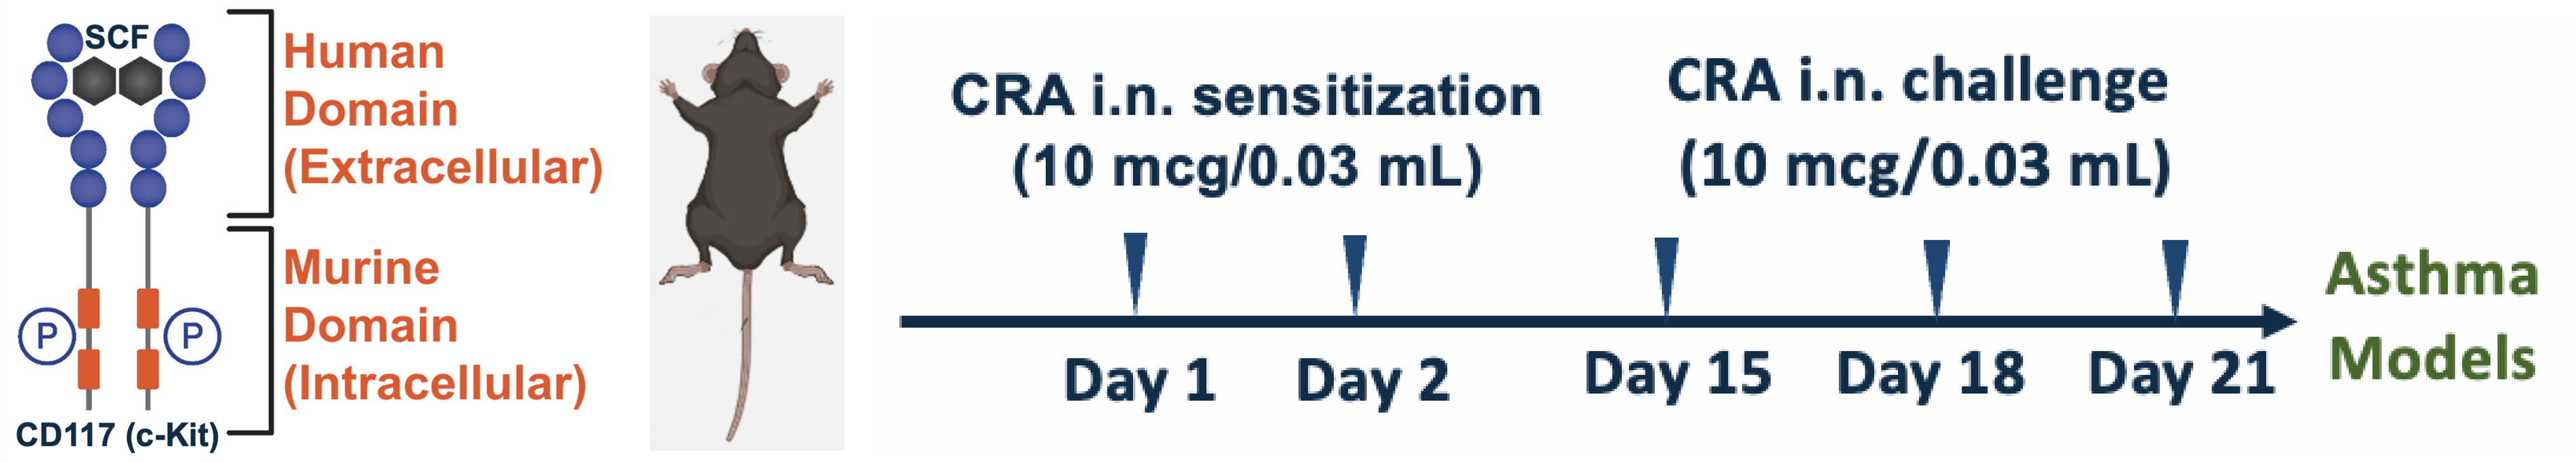
SCF
Human Domain (Extracellular)
Murine Domain (Intracellular)
CD117 (c-Kit)
CRA i.n. sensitization (10 mcg/0.03 mL)
CRA i.n. challenge (10 mcg/0.03 mL)
Day 1
Day 2
Day 15
Day 18
Day 21
Asthma Models

Established Asthma Models

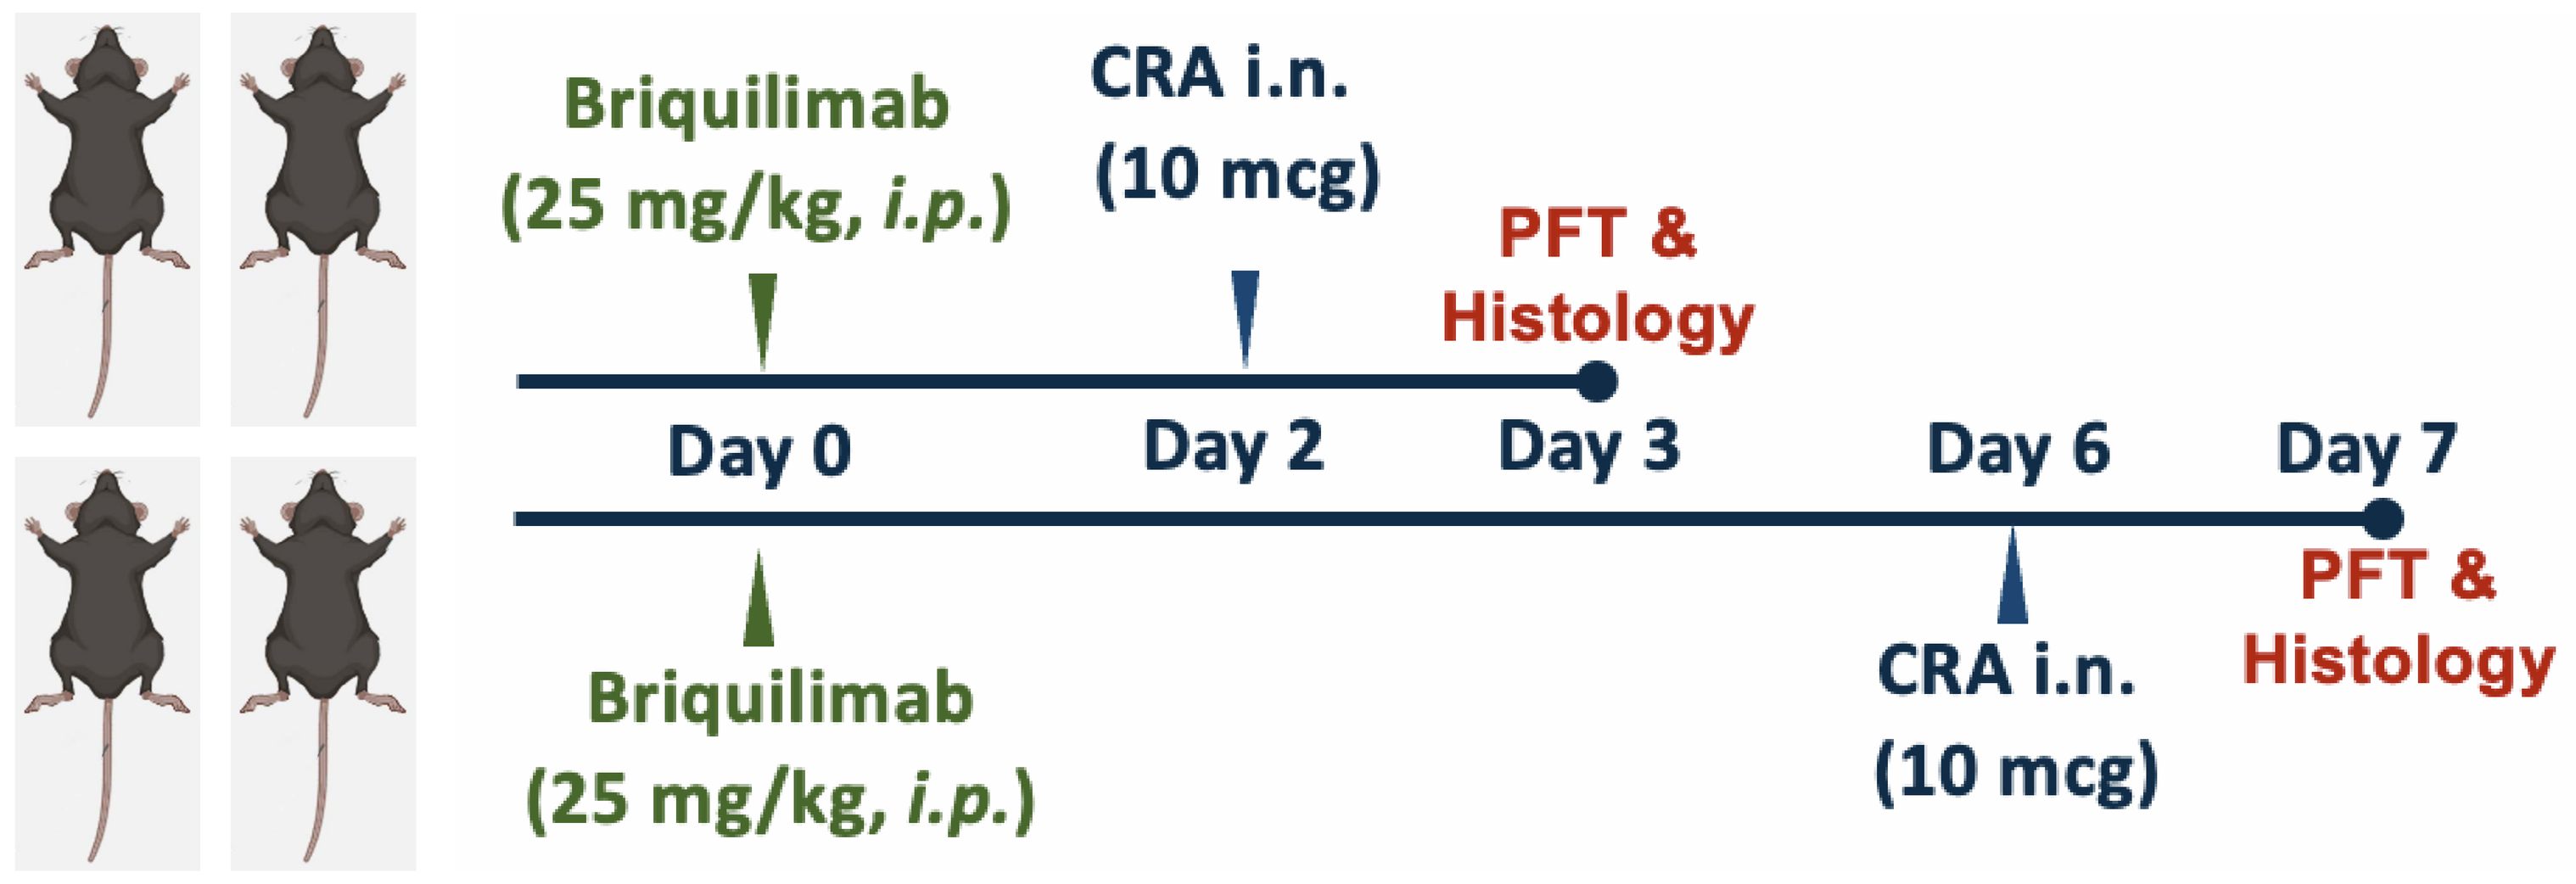
Briquilimab (25 mg/kg, i.p.)
CRA i.n. (10 mcg)
PFT & Histology
Day 0
Day 2
Day 3
Day 6
Day 7
Briquilimab (25 mg/kg, i.p.)
CRA i.n. (10 mcg)
PFT & Histology

PFT: Pulmonary function testing

Three days after one-time therapeutic briquilimab (Briq) treatment, established asthma models exhibited significant decreases in the numbers of mast cells in the lung and the back skin as well as significantly decreased pulmonary resistance and airway inflammation following allergen exposure (Scale bar = 50 mm).

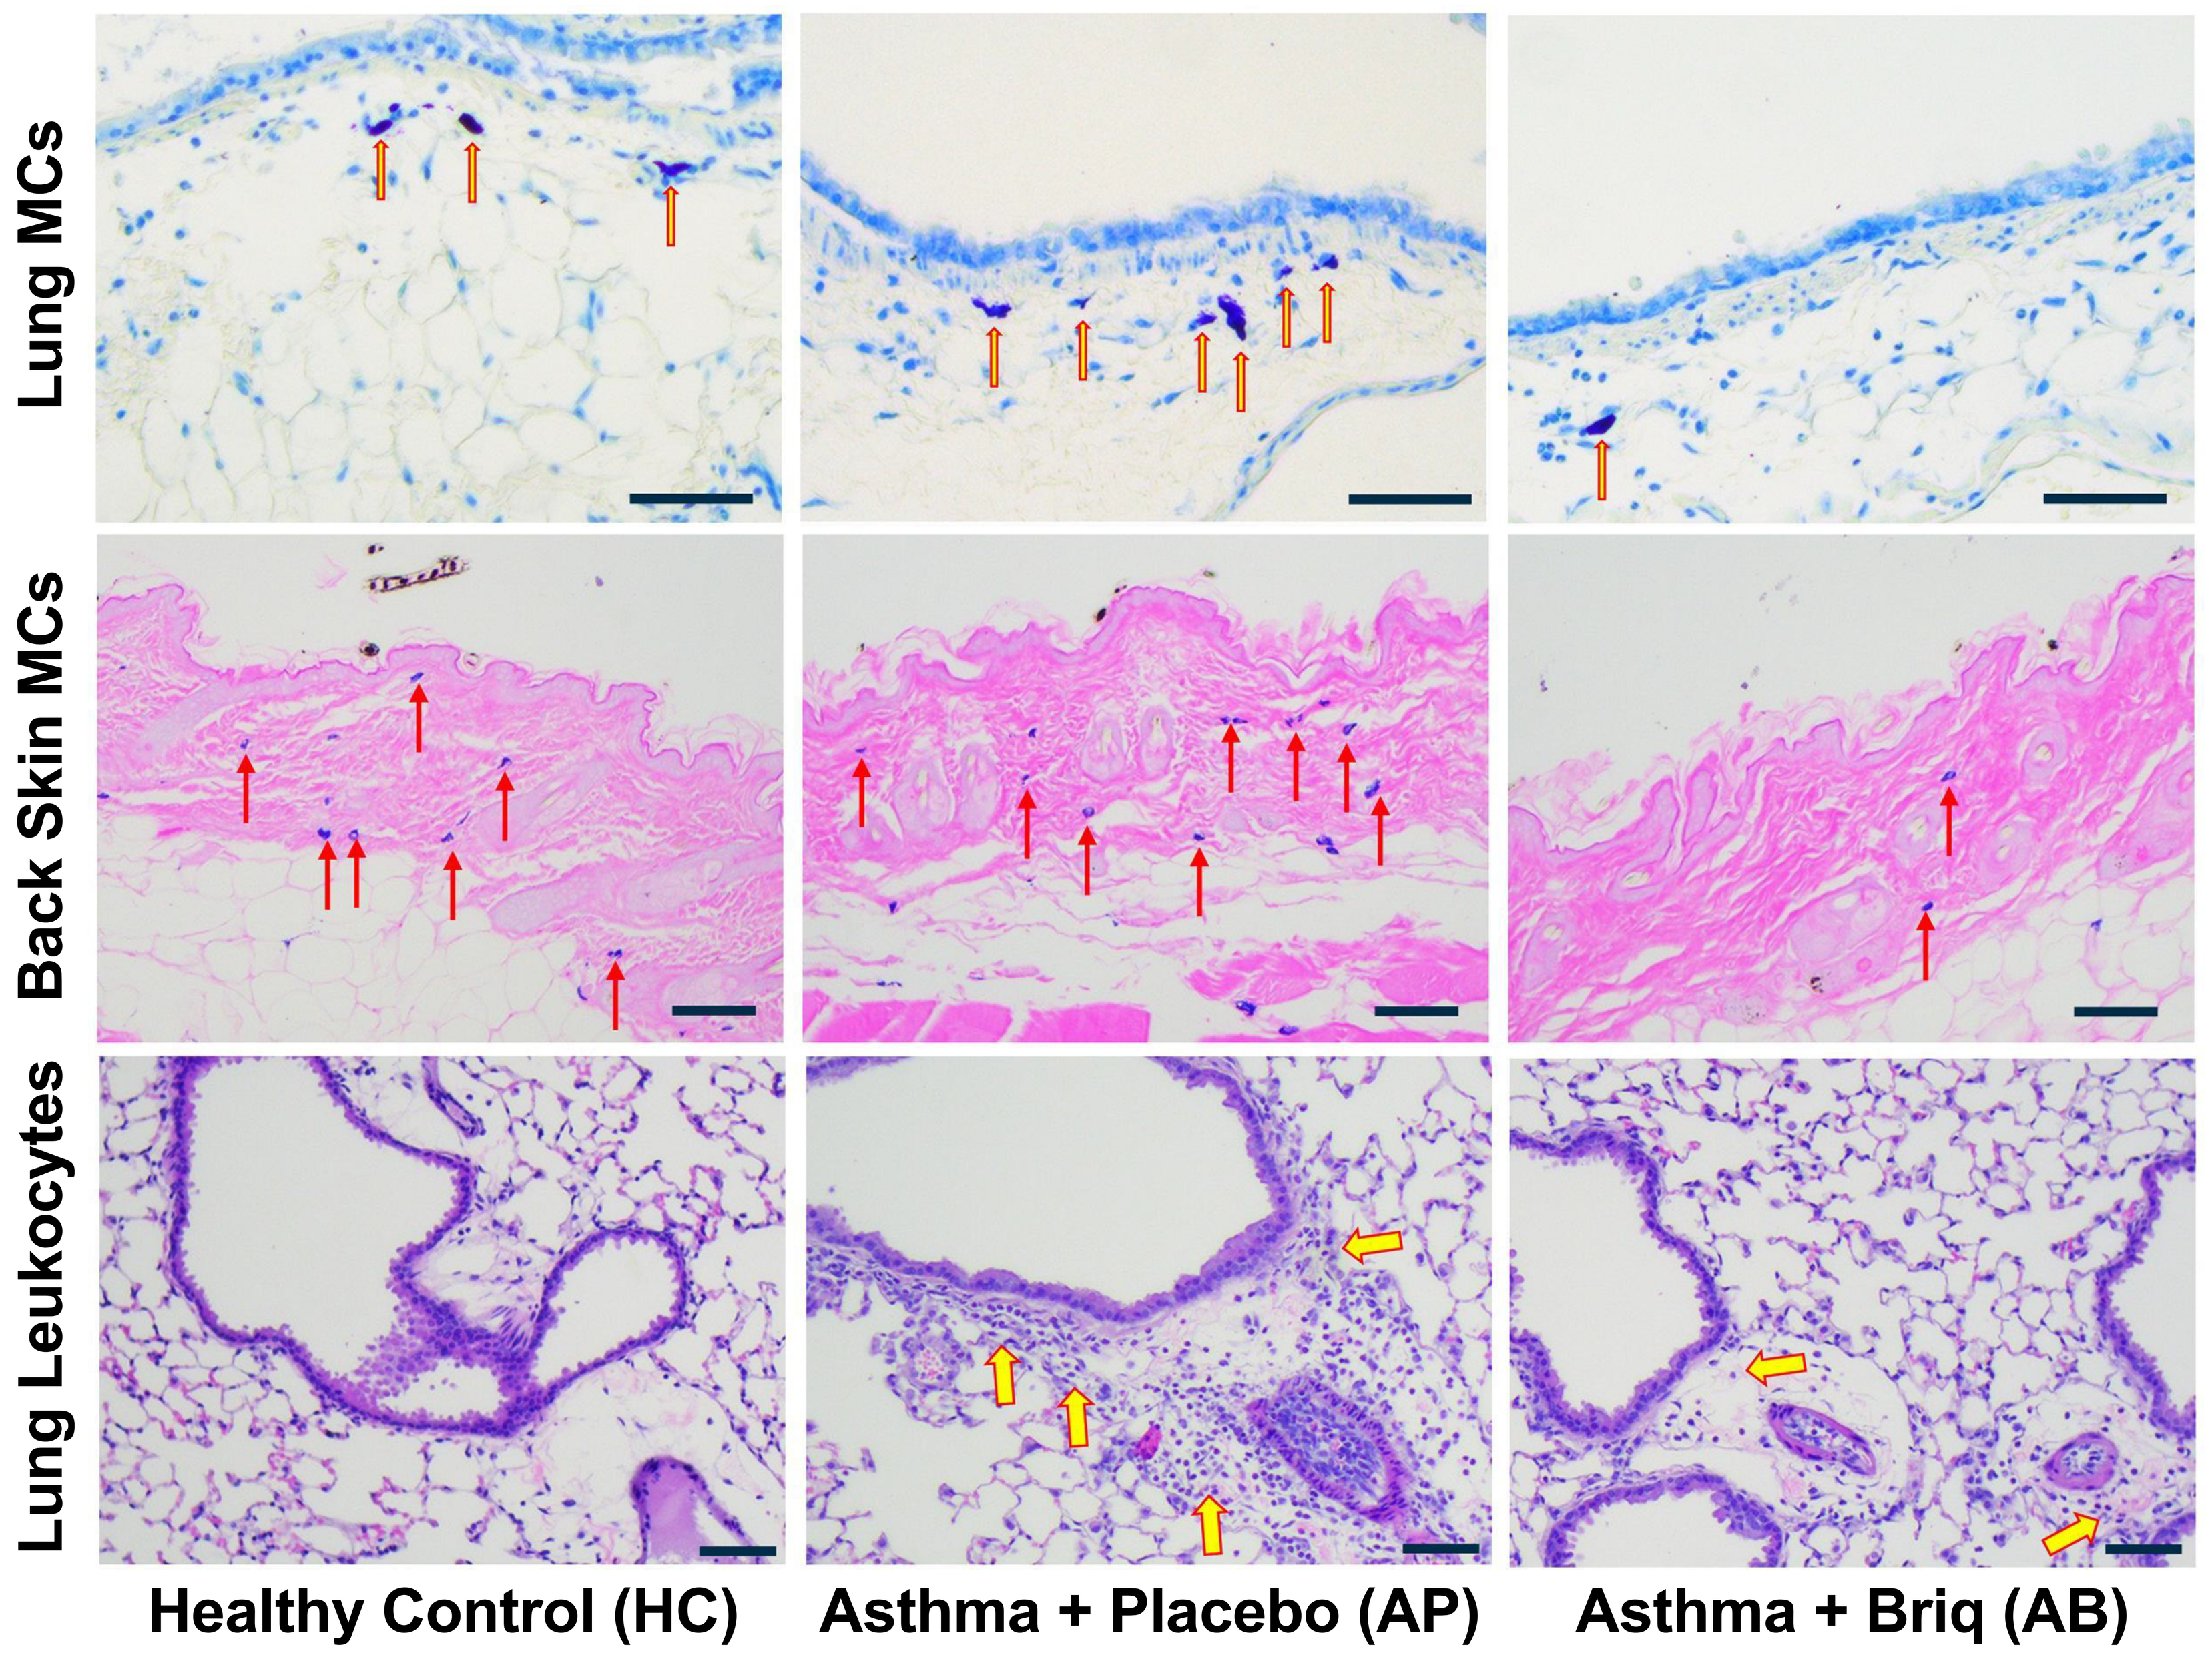
Lung MCs
Back Skin MCs
Lung Leukocytes
Healthy Control (HC)
Asthma + Placebo (AP)
Asthma + Briq (AB)

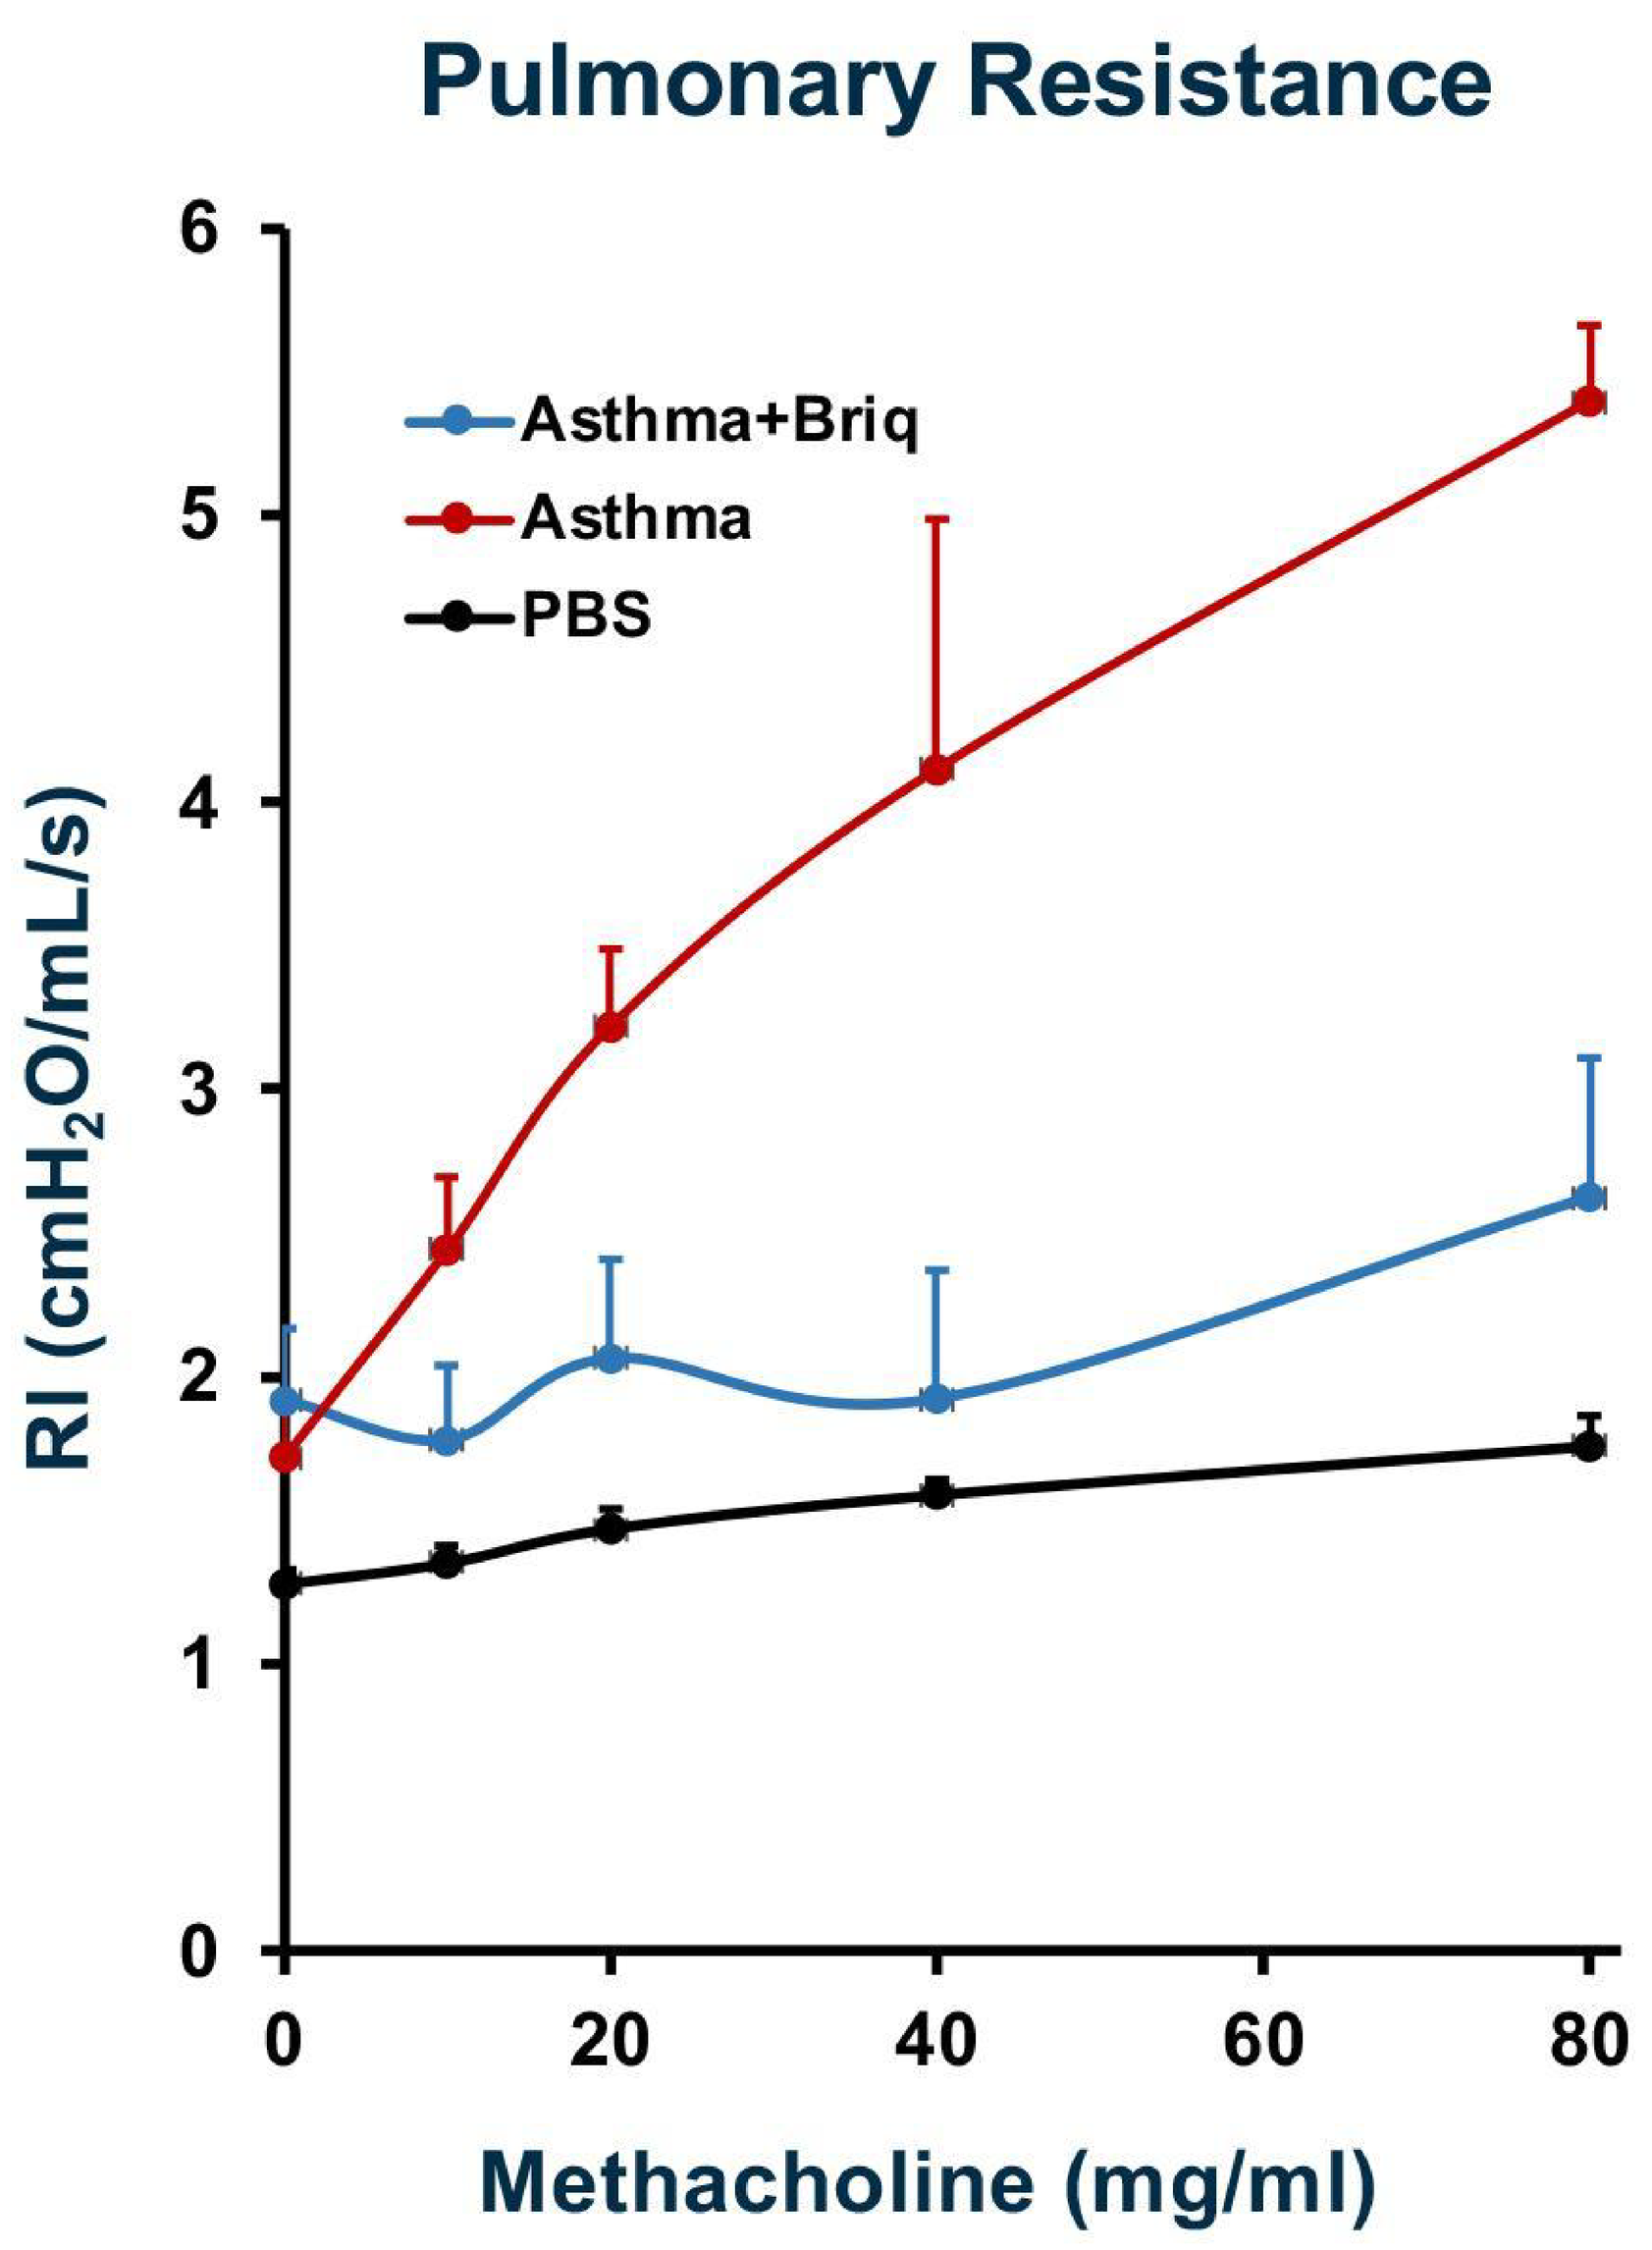
Pulmonary Resistance
Asthma+Briq
Asthma
PBS
RI (cmH2O/mL/s)
Methacholine (mg/ml)

Depletion of mast cells, by briquilimab, may provide a novel therapeutic approach for patients with asthmatic disease.

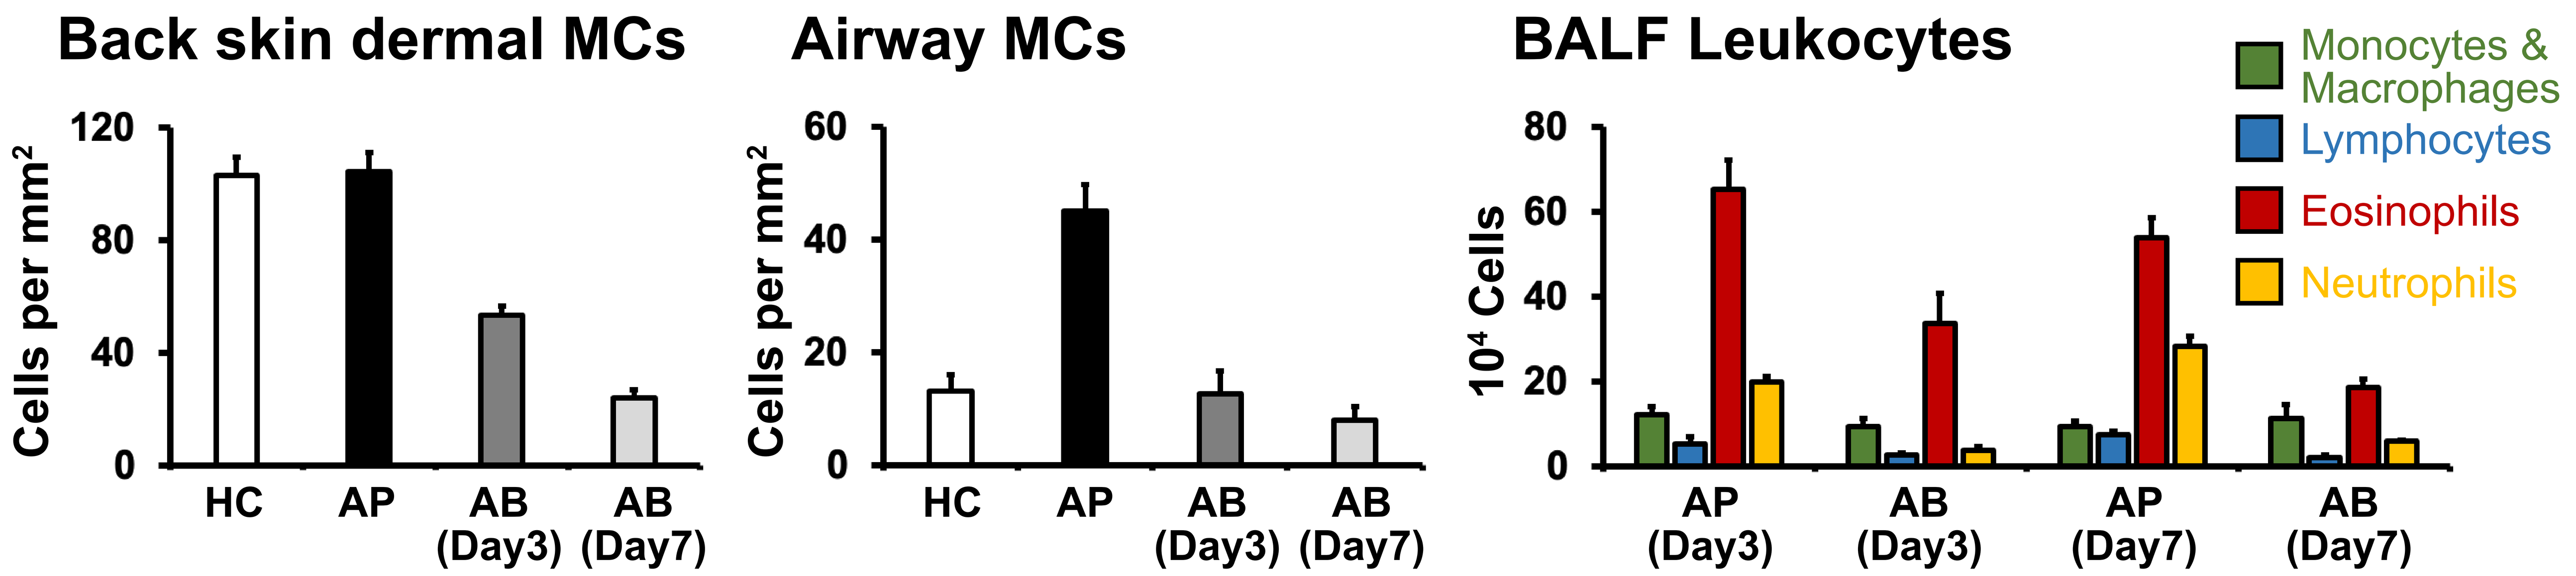
Back skin dermal MCs
Airway MCs
BALF Leukocytes
Cells per mm²
10⁴ Cells
HC
AP
AB (Day3)
AB (Day7)
AP (Day3)
AP (Day7)
Monocytes & Macrophages
Lymphocytes
Eosinophils
Neutrophils

CONCLUSIONS:

- A single dose of briquilimab can deplete mast cells (MCs) in both inflamed (lung) and non-inflamed tissues (back skin) of an asthma model elicited in h/mCD117 mice.
- Briquilimab's ability to deplete MCs contributes to its therapeutic effect on asthma models elicited in h/mCD117 mice.

TAKEAWAYS:

- Mast cells play a key role in asthma pathogenesis.
- Briquilimab can potentially target MCs in the treatment for allergic asthma.

ACKNOWLEDGEMENTS:

- Drs. Stephen Galli & Judith Shizuru, Stanford University.
- Edwin Tucker, Daniel Adelman, Patricia Carlos, Ryan Taylor, Aaron Miller, Ameer Alameddin, Kavitha Arulprakasam, and Anne Le, Jasper Therapeutics, Inc.

Briquilimab is an investigational product and not approved for any indication.